Presented at the
25th Anniversary Congress on Women's Health
April 28–30, 2017 • Washington, DC

Efficacy of Bremelanotide Among Women Completing the Core Phase of the RECONNECT Studies

Sheryl Kingsberg,[1] Robin Kroll,[2] Johna Lucas,[3] Robert Jordan,[3] Carl Spana[3]

[1]Case Western Reserve University School of Medicine, Cleveland, OH, USA; [2]Seattle Women's Health, Research, Gynecology, Seattle, WA, USA; [3]Palatin Technologies, Inc, Cranbury, NJ, USA

Background

- Female sexual dysfunctions (FSD) comprise a group of conditions with physiological, psychological, and social components[1]
- The most common sexual concern expressed by women with FSD is diminished or lack of desire for sexual activity. When accompanied by distress, this may be diagnosed as hypoactive sexual desire disorder (HSDD)[2]
- Bremelanotide (BMT; PT-141) is an investigational, novel cyclic 7-amino agonist with high affinity for the type-4 receptor[3]
- As an analog of the naturally occurring peptide α-melanocyte-stimulating hormone, BMT acts on the physiological and neurobiological components of female sexual function, with the potential to modulate brain pathways involved in sexual arousal and desire in women with HSDD[4]
- BMT is currently being evaluated for the treatment of HSDD (with or without decreased arousal) in premenopausal women

Study Design

- The RECONNECT studies comprise 2, identical, randomized, Phase 3, placebo controlled, multicenter trials (NCT02333071 [Study 301] and NCT02338960 [Study 302])
- The Core phase of the trials includes a 1-month no-drug qualification period, a 1-month single-blind placebo treatment period (to establish baseline), and a 24-week double-blind treatment period. Both trials also have an ongoing 52-week open label extension (**Figure 1**)

Figure 1. Study Design

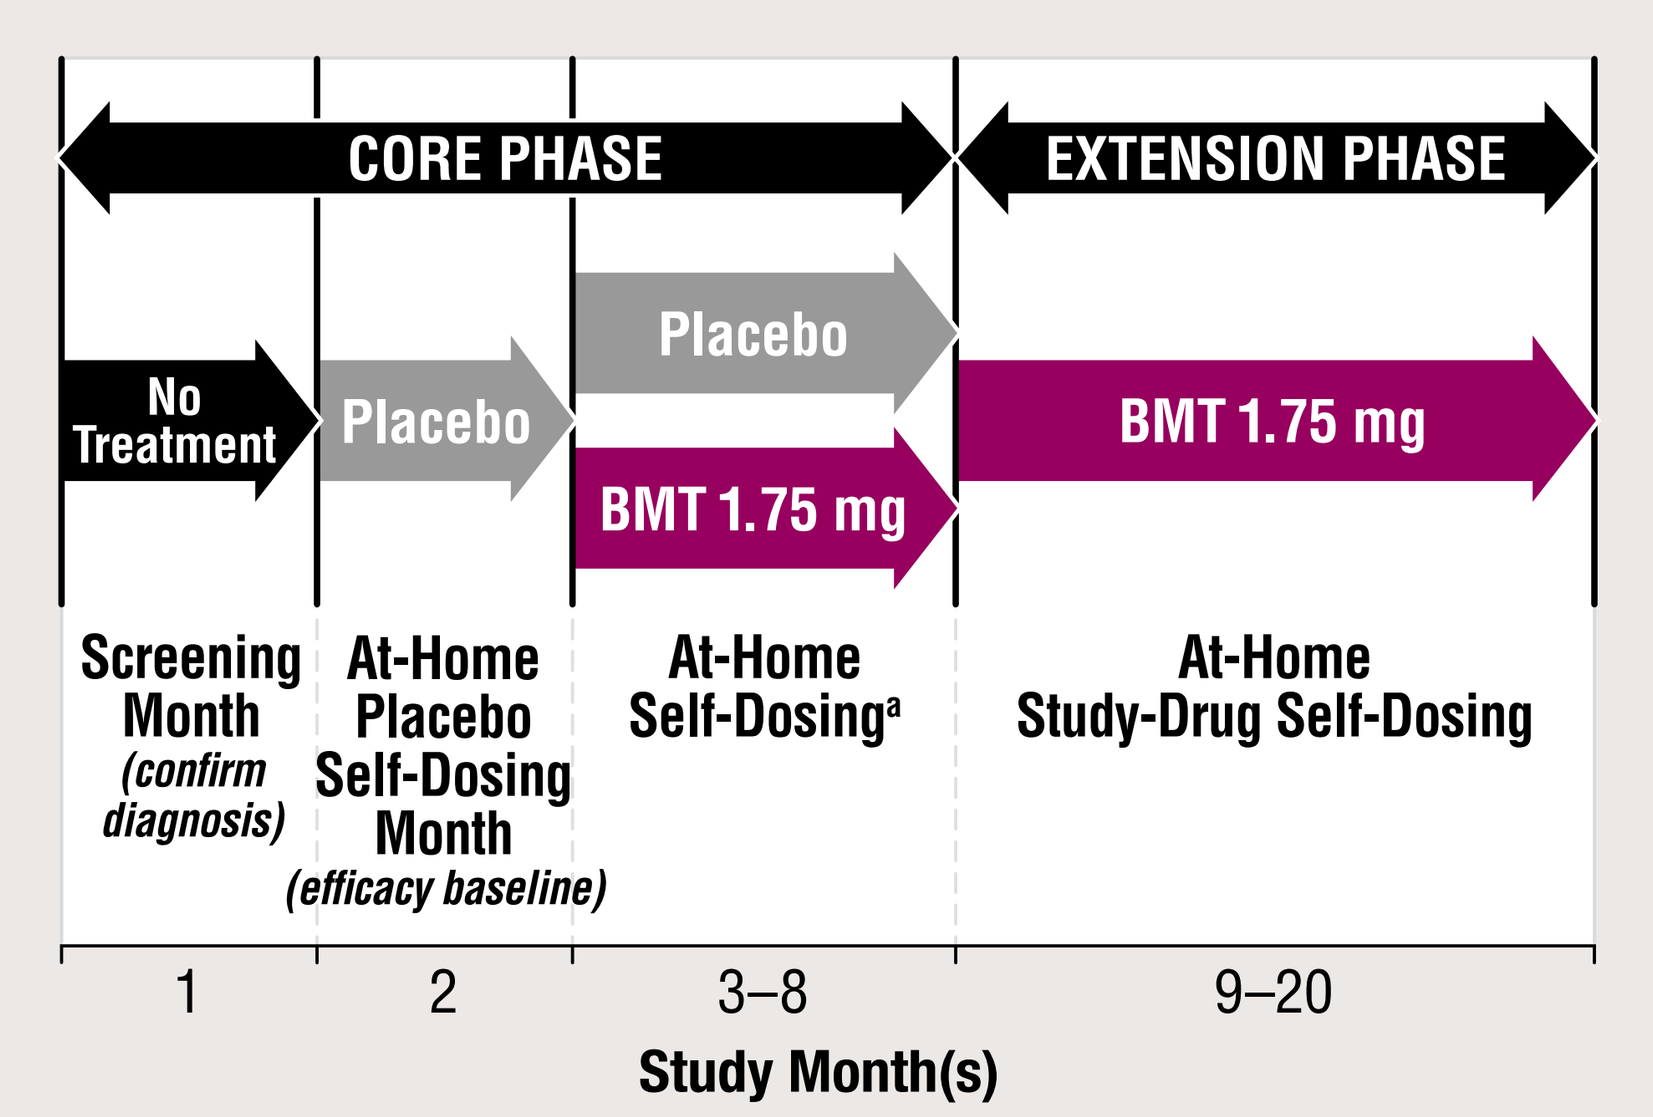

CORE PHASE
EXTENSION PHASE
No Treatment
Placebo
Placebo
BMT 1.75 mg
BMT 1.75 mg
Screening Month (confirm diagnosis)
At-Home Placebo Self-Dosing Month (efficacy baseline)
At-Home Self-Dosing[a]
At-Home Study-Drug Self-Dosing
1
2
3–8
9–20
Study Month(s)

[a]Participants self-administered BMT 1.75 mg or placebo subcutaneously using an auto-injector, as-desired, prior to sexual activity.
BMT, bremelanotide.

- Participants were randomized (1:1) to either placebo or BMT 1.75 mg, self-administered, subcutaneously using an auto-injector (**Figure 2**), as desired, prior to sexual activity

Figure 2. Auto-Injector

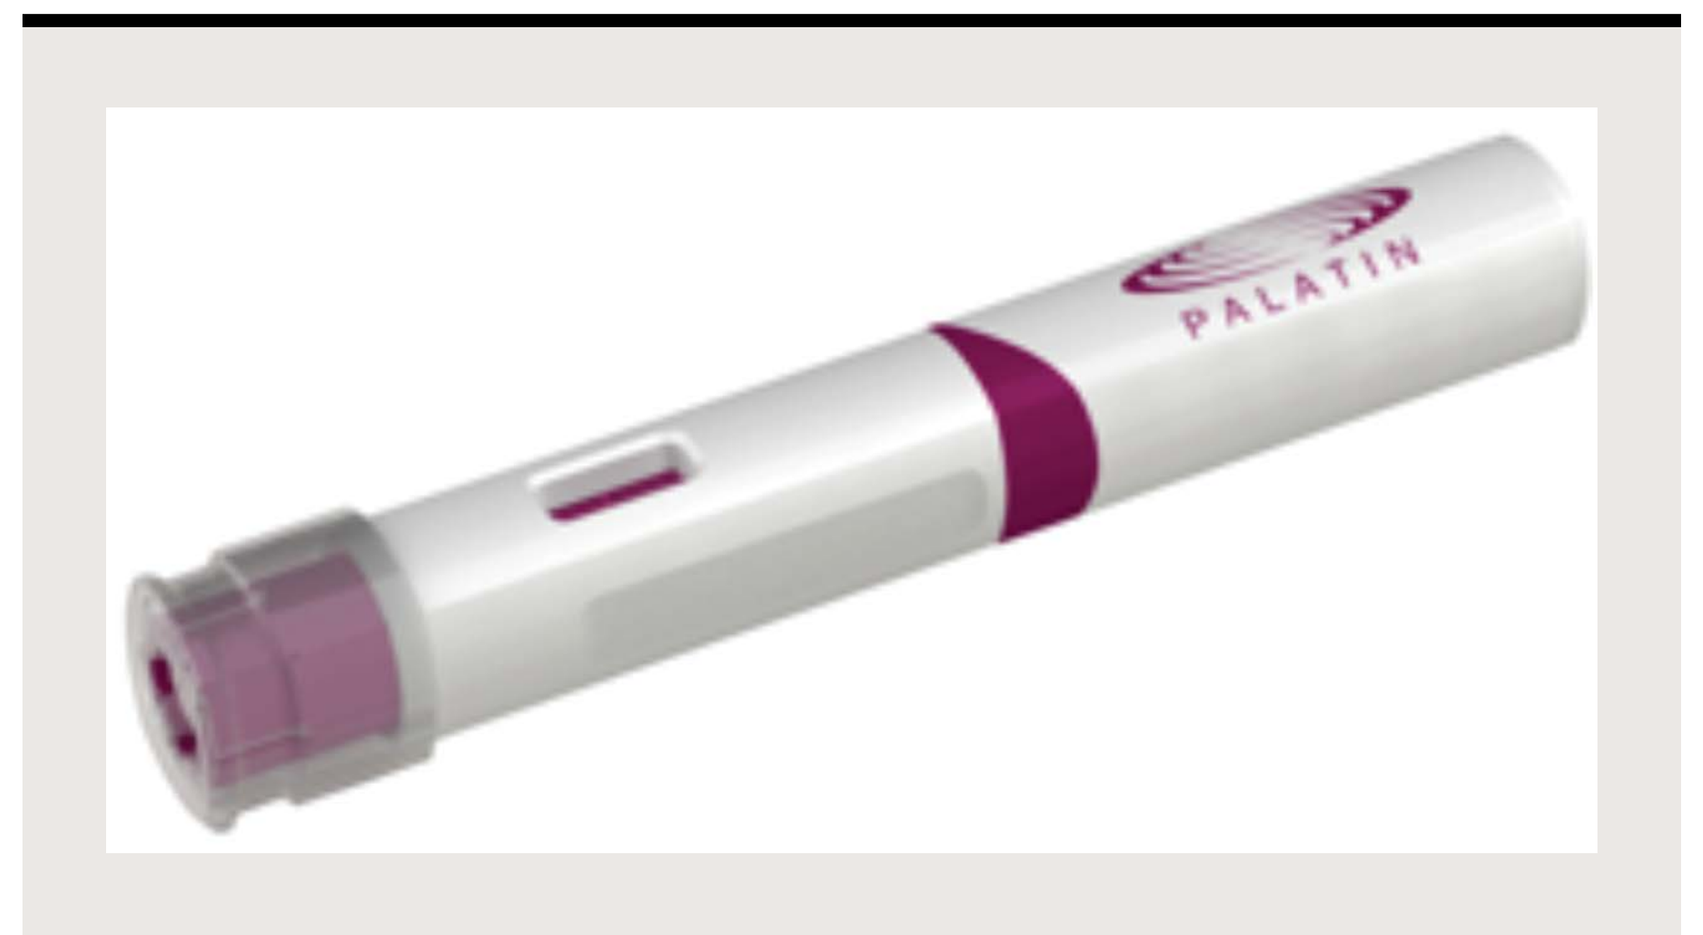

PALATIN

Study Objectives

- To evaluate the safety and efficacy of BMT 1.75 mg self-administered, as desired, for the treatment of HSDD

Study Population

- Healthy, premenopausal, nonpregnant women, ≥18 years of age, currently in a stable (≥6 months) relationship
- Diagnosed with HSDD (with or without decreased arousal) for ≥6 months
- Experienced 'normal' sexual function at some point in the past for ≥2 years
- Willing to engage in sexual activities ≥1×/month during the study
- Had ***ALL*** of the following at screening:
 - —Patient Health Questionnaire-9 (PHQ-9) total score <10, and score of 0 on question 9
 - —Female Sexual Function Index (FSFI) total score ≤26 (if diagnosed with HSDD with/without symptoms of decreased arousal), ***OR***
 - —FSFI desire domain score ≤5 (if diagnosed with HSDD without decreased arousal) regardless of total FSFI score
 - —Female Sexual Distress Scale-Desire/Arousal/Orgasm (FSDS-DAO) total score >18

Assessments

- The FSFI is a 19-item measure of female sexual function[5,6] consisting of 6 domains: Desire, Arousal, Lubrication, Orgasm, Satisfaction, and Pain
 - —Scores range from 0 to 6 for Arousal, Lubrication, Orgasm, and Pain; from 1.2 to 6 for Desire; and from 0.8 to 6 for Satisfaction
 - —The total score is the sum of the domain scores and ranges from 2 to 36
 - —The recall period is the past 4 weeks
 - —Higher scores on indicate a greater level of sexual function
- The FSDS-DAO is a 15-item instrument based on the 13-item Female Sexual Distress Scale-Revised (FSDS-R);[7] both questionnaires are designed to evaluate different aspects of sexual-related distress over the past 30 days
 - —Item 13 relates specifically to 'bother' related to sexual desire
 - —Responses are provided using Likert-type scale and range from 0 (never) to 4 (always)
 - —The total score is calculated as the sum of the responses and ranges from 0 to 60
 - —Higher scores on the FSDS-DAO indicates greater sexual-related distress

Key Outcome Measures

Coprimary Efficacy Endpoints

- Change from baseline to end-of-study (EOS) in the FSFI desire domain score
- Change from baseline to EOS in the score for feeling bothered by low sexual desire as measured by Item 13 of the FSDS-DAO

Responder Analysis Based On

- Participants self-reporting a score of ≥5 (on a 7-point Likert scale) in response to question 3 on the General Assessment Questionnaire (GAQ) "To what degree do you think you benefited from taking the study drug?"
- The proportion of participants meeting or exceeding the following predefined minimal clinically important differences (MCIDs)
 - —FSFI-D score (MCID=0.6)
 - —FSDS-DAO Item 13 score (MCID=−1.0)

Results

The Core phase of both studies is complete; the open-label Extension phase is ongoing

Participants

- 1267 women were randomized; 1247 women comprise the safety population; 1202 comprise the primary efficacy population (mITT)
- A total of 856 women completed the double-blind, Core phase of the study
- The disposition of subjects is shown in **Figure 3**

Figure 3. Subject Disposition

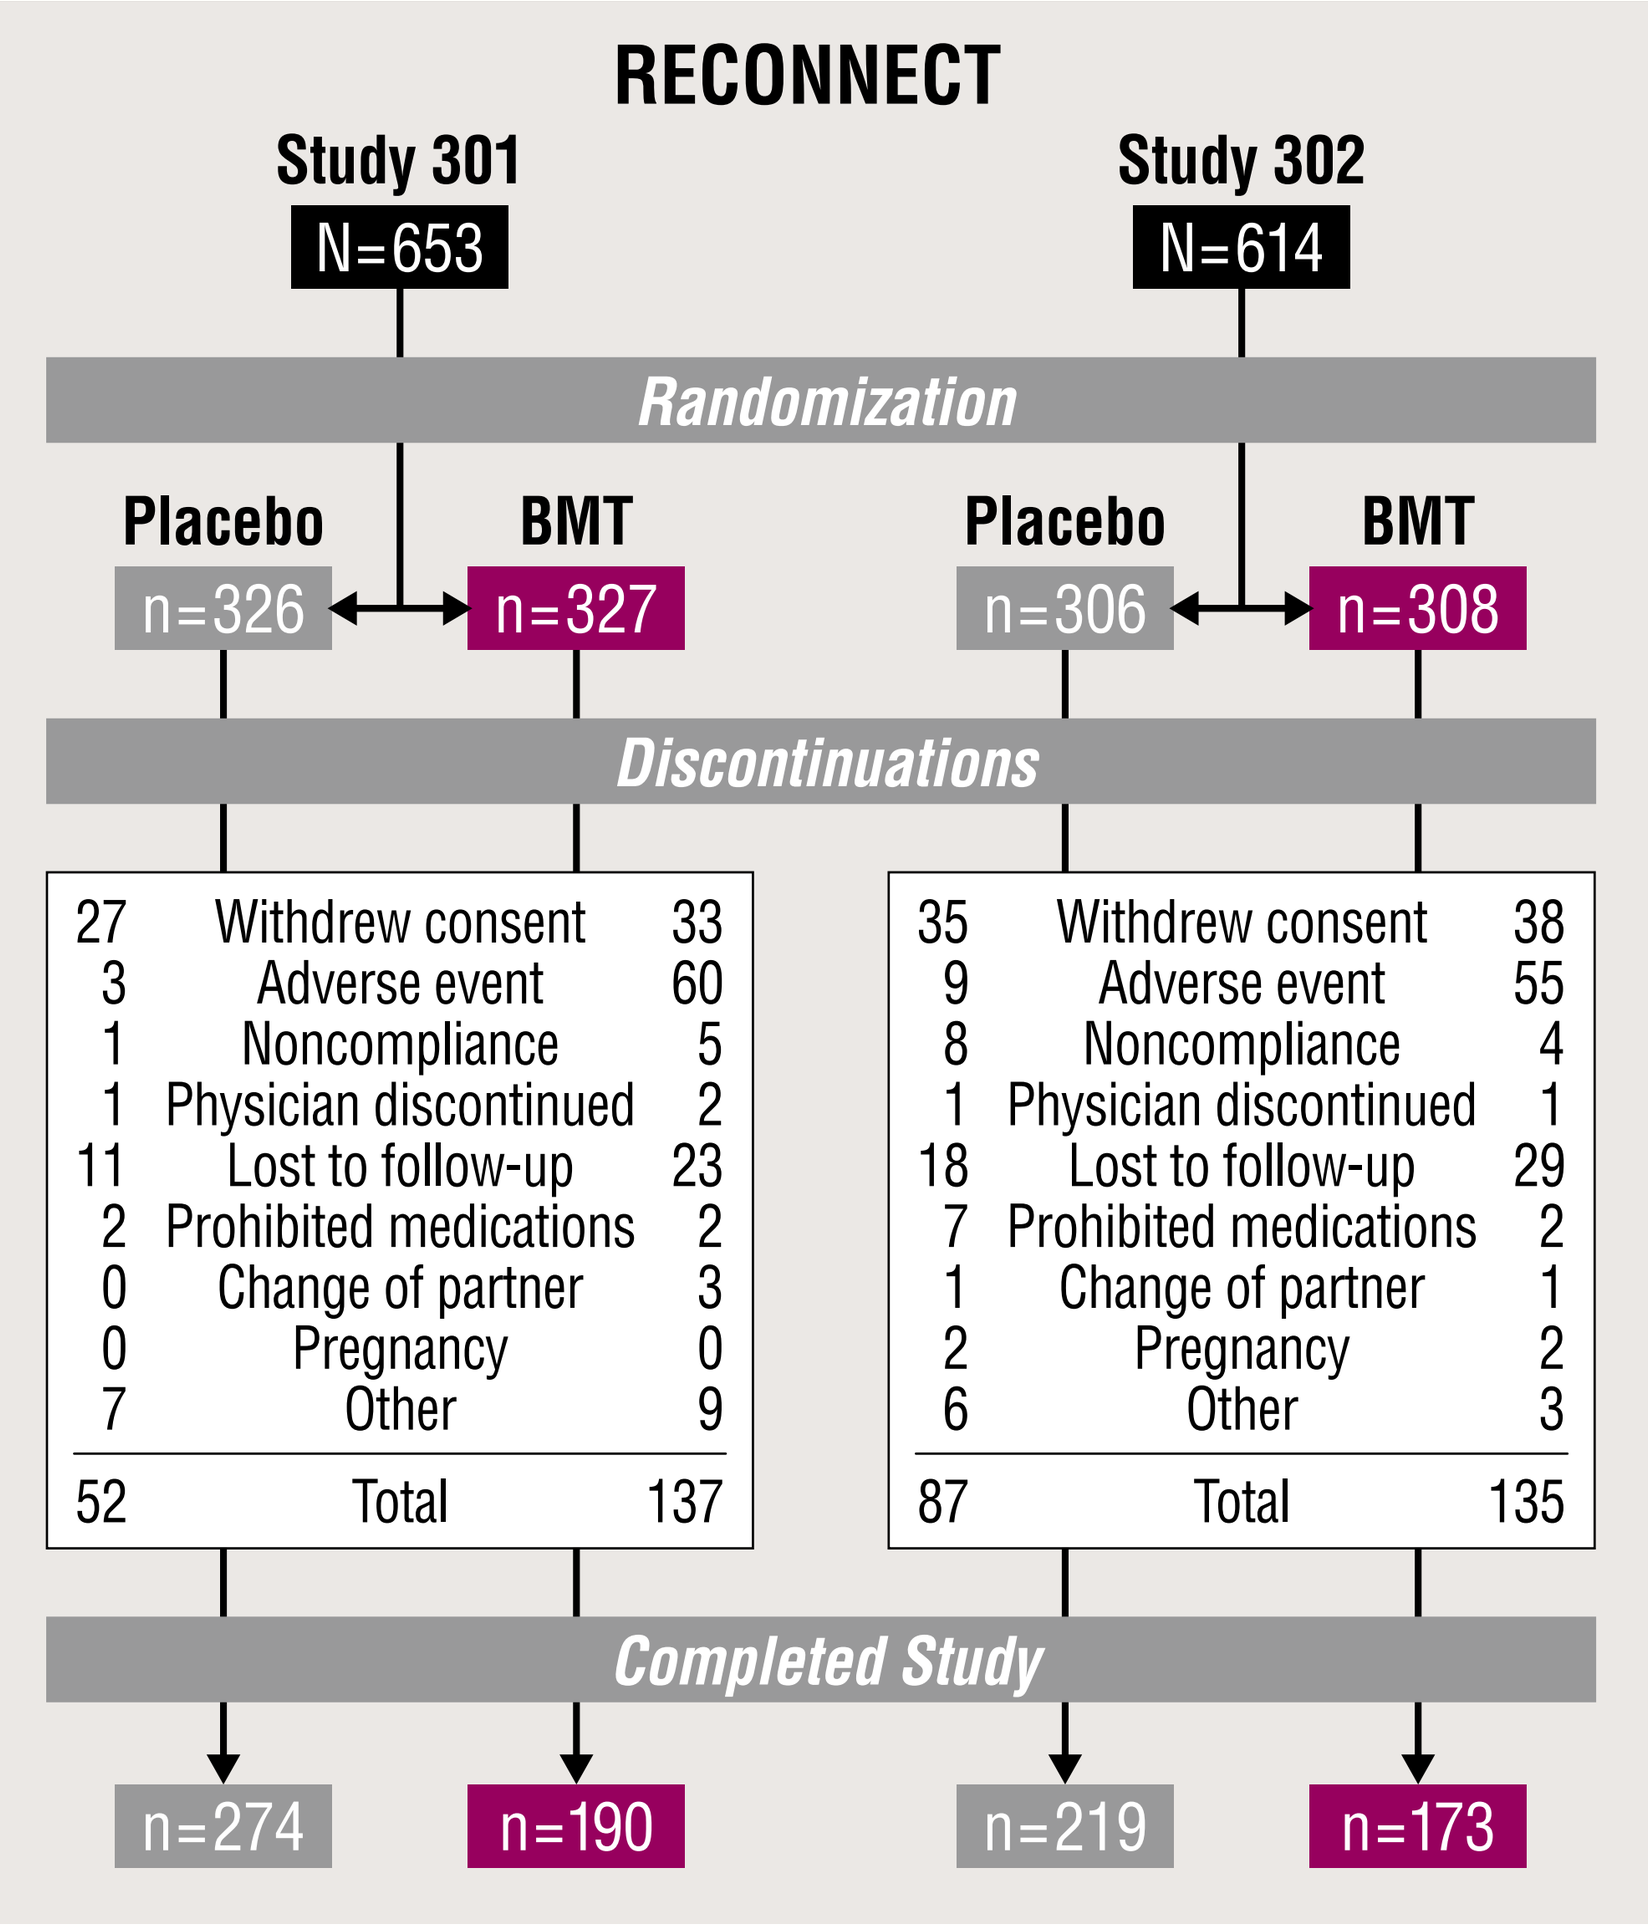

RECONNECT
Study 301
N=653
Study 302
N=614
Randomization
Placebo n=326
BMT n=327
Placebo n=306
BMT n=308
Discontinuations
27 Withdrew consent 33
3 Adverse event 60
1 Noncompliance 5
1 Physician discontinued 2
11 Lost to follow-up 23
2 Prohibited medications 2
0 Change of partner 3
0 Pregnancy 0
7 Other 9
52 Total 137
35 Withdrew consent 38
9 Adverse event 55
8 Noncompliance 4
1 Physician discontinued 1
18 Lost to follow-up 29
7 Prohibited medications 2
1 Change of partner 1
2 Pregnancy 2
6 Other 3
87 Total 135
Completed Study
n=274
n=190
n=219
n=173

BMT, bremelanotide.

- Participants were mostly white (84.3% Study 301; 86.9% Study 302) and non-Hispanic/Latina (>90% in both studies)
- The most frequent diagnosis was HSDD with decreased arousal (mean time since diagnosis ≥45 months)
- Baseline demographics are shown in **Table 1**

Table 1. Demographic Characteristics at Screening (Safety Population)

Variable	301			302		
	Placebo (n=319)	BMT (n=324)	Total (N=643)	Placebo (n=301)	BMT (n=303)	Total (N=604)
Mean age (SD), y	38.5 (7.2)	38.4 (7.0)	38.5 (7.1)	39.1 (7.0)	38.5 (7.2)	38.8 (7.1)
Mean weight (SD), kg	76.9 (19.6)	78.8 (20.4)	77.9 (20.0)	76.9 (18.2)	78.2 (19.3)	77.6 (18.8)
Mean height (SD), m	1.64 (0.08)	1.65 (0.07)	1.65 (0.07)	1.65 (0.07)	1.65 (0.07)	1.65 (0.07)
Mean BMI (SD), kg/m²	28.5 (7.3)	28.9 (7.0)	28.7 (7.2)	28.4 (6.5)	28.8 (7.0)	28.6 (6.8)
HSDD Diagnosis, n (%)						
With decreased arousal	240 (75)	238 (74)	478 (74)	206 (68)	205 (68)	411 (68)
Without decreased arousal	79 (25)	86 (27)	1654 (26)	95 (32)	98 (32)	193 (32)
Months (SD) since HSDD diagnosis	49.0 (43.7)	48.3 (42.2)	48.6 (42.9)	45.8 (43.6)	43.7 (42.2)	44.8 (42.9)

BMI, body mass index; BMT, bremelanotide; HSDD, hypoactive sexual desire disorder; SD, standard deviation.

Efficacy Results: FSFI-D (Completers)

- Compared with those taking placebo, the FSFI-D score increased in women using BMT 1.75 mg from the first month of double-blind treatment (**Figure 4**)
- Following a sensitivity analysis that assumed all dropouts were treatment failures, the effect size decreased but results still showed statistically significant improvement in comparison to placebo

Figure 4. Mean FSFI-D Scores for Placebo and BMT Over the Core Phase

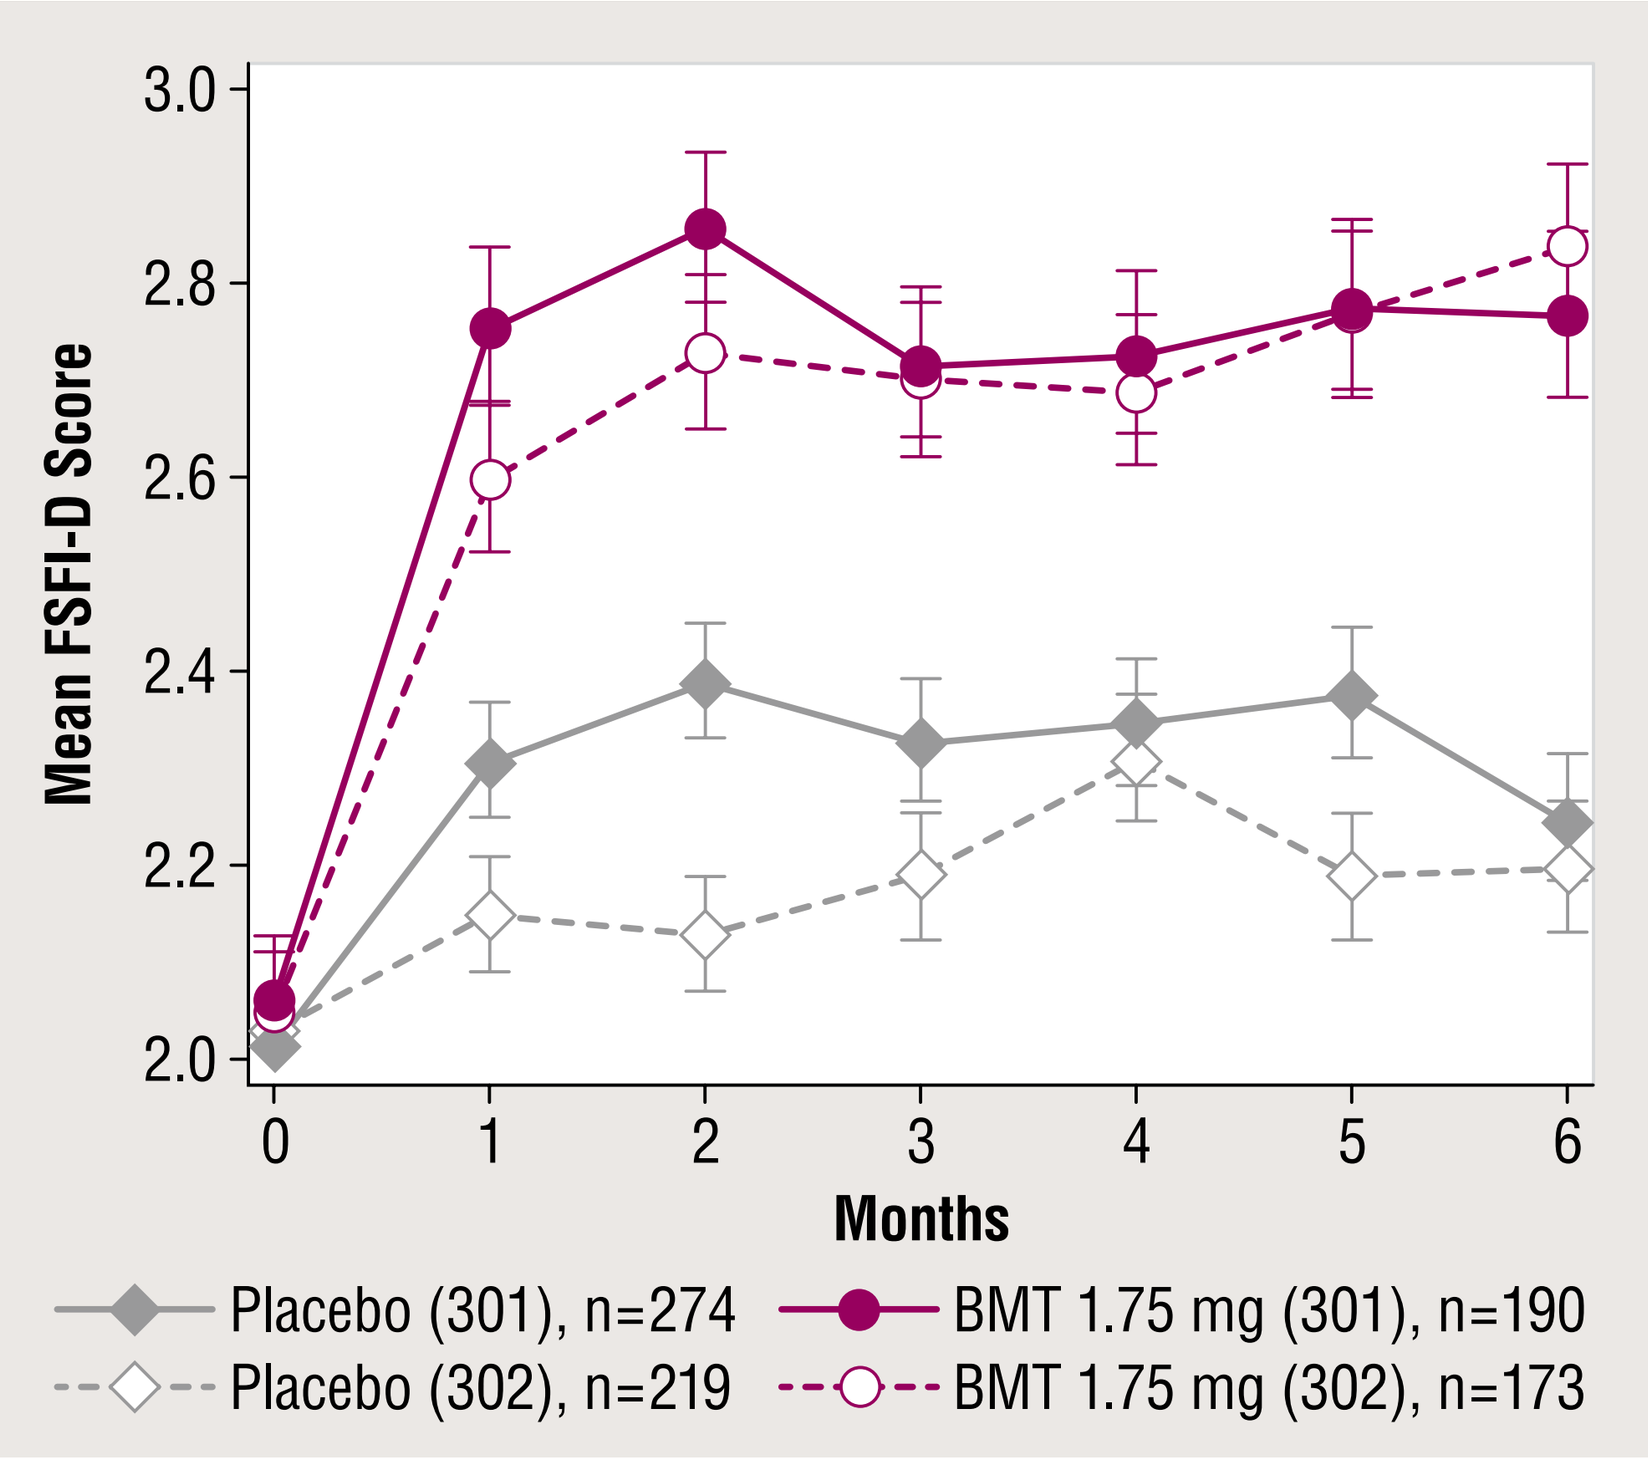

Mean FSFI-D Score
3.0
2.8
2.6
2.4
2.2
2.0
0
1
2
3
4
5
6
Months
Placebo (301), n=274
BMT 1.75 mg (301), n=190
Placebo (302), n=219
BMT 1.75 mg (302), n=173

Error bars are standard error of the mean.
BMT, bremelanotide; FSFI-D, Female Sexual Function Index Desire domain.

- Compared with those taking placebo, women taking BMT had significantly increased scores on the desire domain of the FSFI at 6 months, indicating an increase in desire (**Figure 5**)

Figure 5. Change in FSFI-D from Baseline to End of Core (Double-Blind) Phase

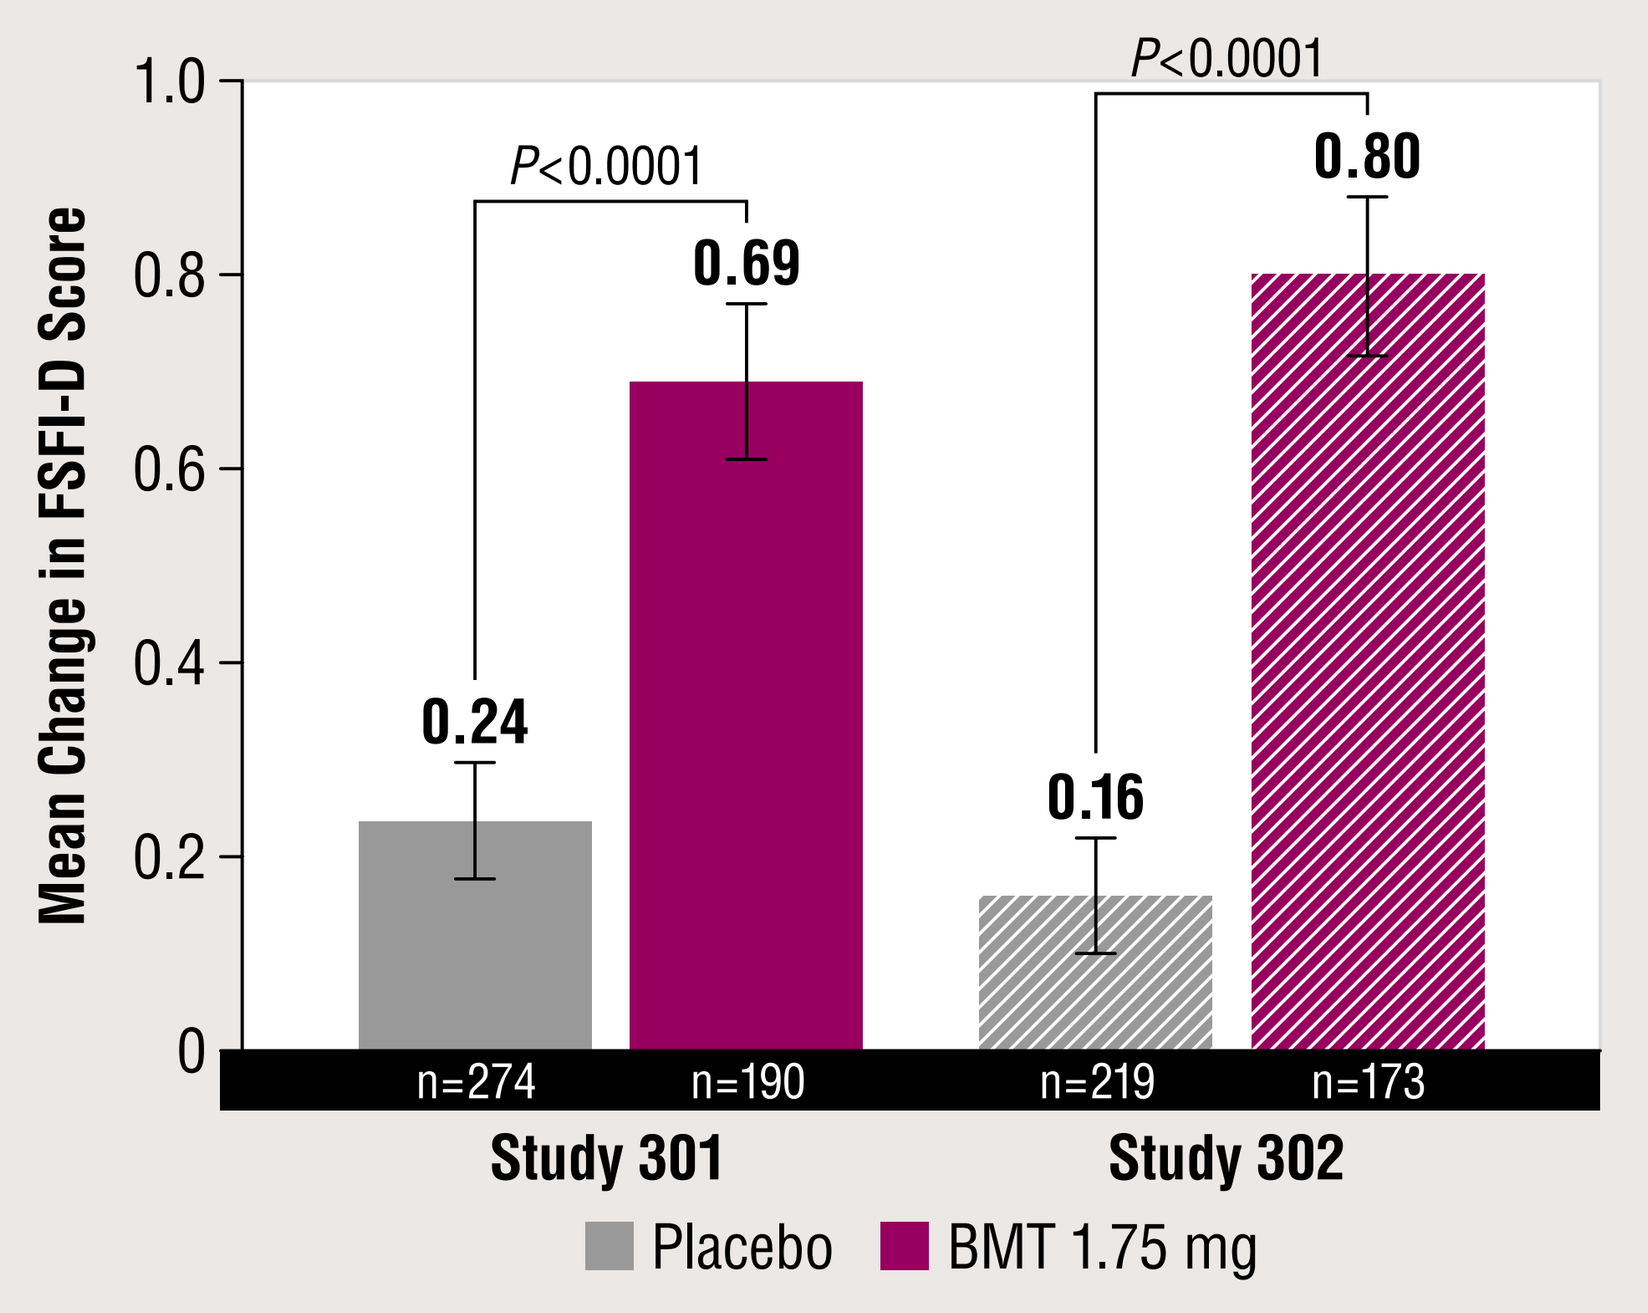

Mean Change in FSFI-D Score
1.0
0.8
0.6
0.4
0.2
0
P<0.0001
0.24
0.69
P<0.0001
0.16
0.80
n=274
n=190
n=219
n=173
Study 301
Study 302
Placebo
BMT 1.75 mg

BMT, bremelanotide; FSFI-D, Female Sexual Function Index Desire domain.
P-values determined by unadjusted Wilcoxon rank-sum test; error bars are standard error of the mean.

Efficacy Results: FSFI-D (Completers)

- Relative to women taking placebo, FSDS-DAO item 13 scores decreased among women taking BMT 1.75 mg from the first month of double-blind treatment (**Figure 6**)

Figure 6. Mean FSDS-DAO Item 13* Scores for Placebo and BMT Over the Core Phase

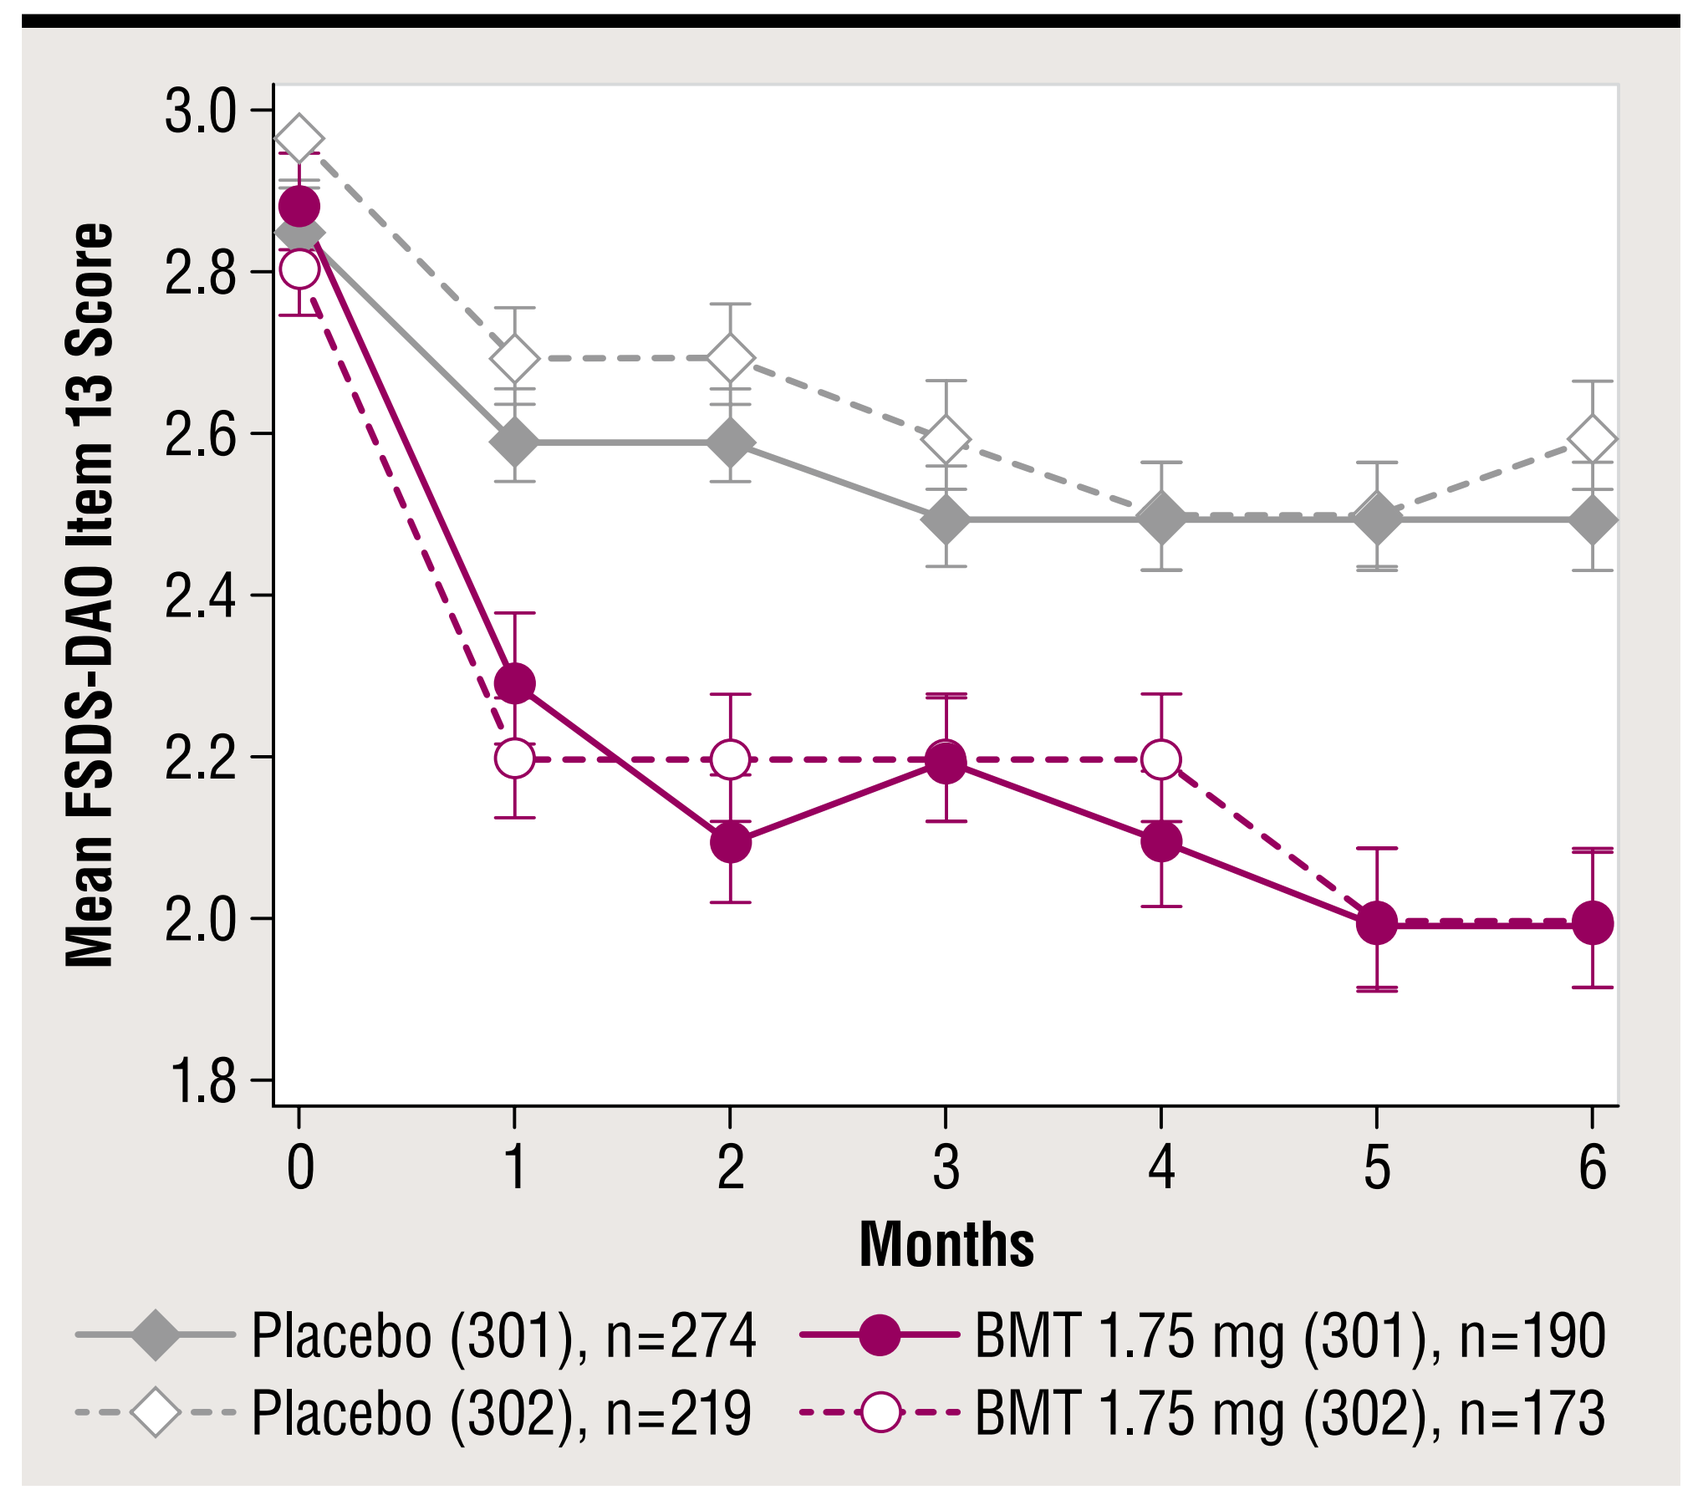

Mean FSDS-DAO Item 13 Score
3.0
2.8
2.6
2.4
2.2
2.0
1.8
0
1
2
3
4
5
6
Months
Placebo (301), n=274
BMT 1.75 mg (301), n=190
Placebo (302), n=219
BMT 1.75 mg (302), n=173

Error bars are standard error of the mean.
*ie, How often do you feel bothered by low sexual desire?
BMT, bremelanotide; FSDS-DAO, Female Sexual Distress Scale–Desire/Arousal/Orgasm.

- Compared with those taking placebo, women using BMT had a significant reduction in their FSDS-DAO Item 13 score at 6 months, indicating a reduction in distress related to low sexual desire (**Figure 7**)

Figure 7. Change in FSDS-DAO Item 13* from Baseline to End of Core (Double-Blind) Phase

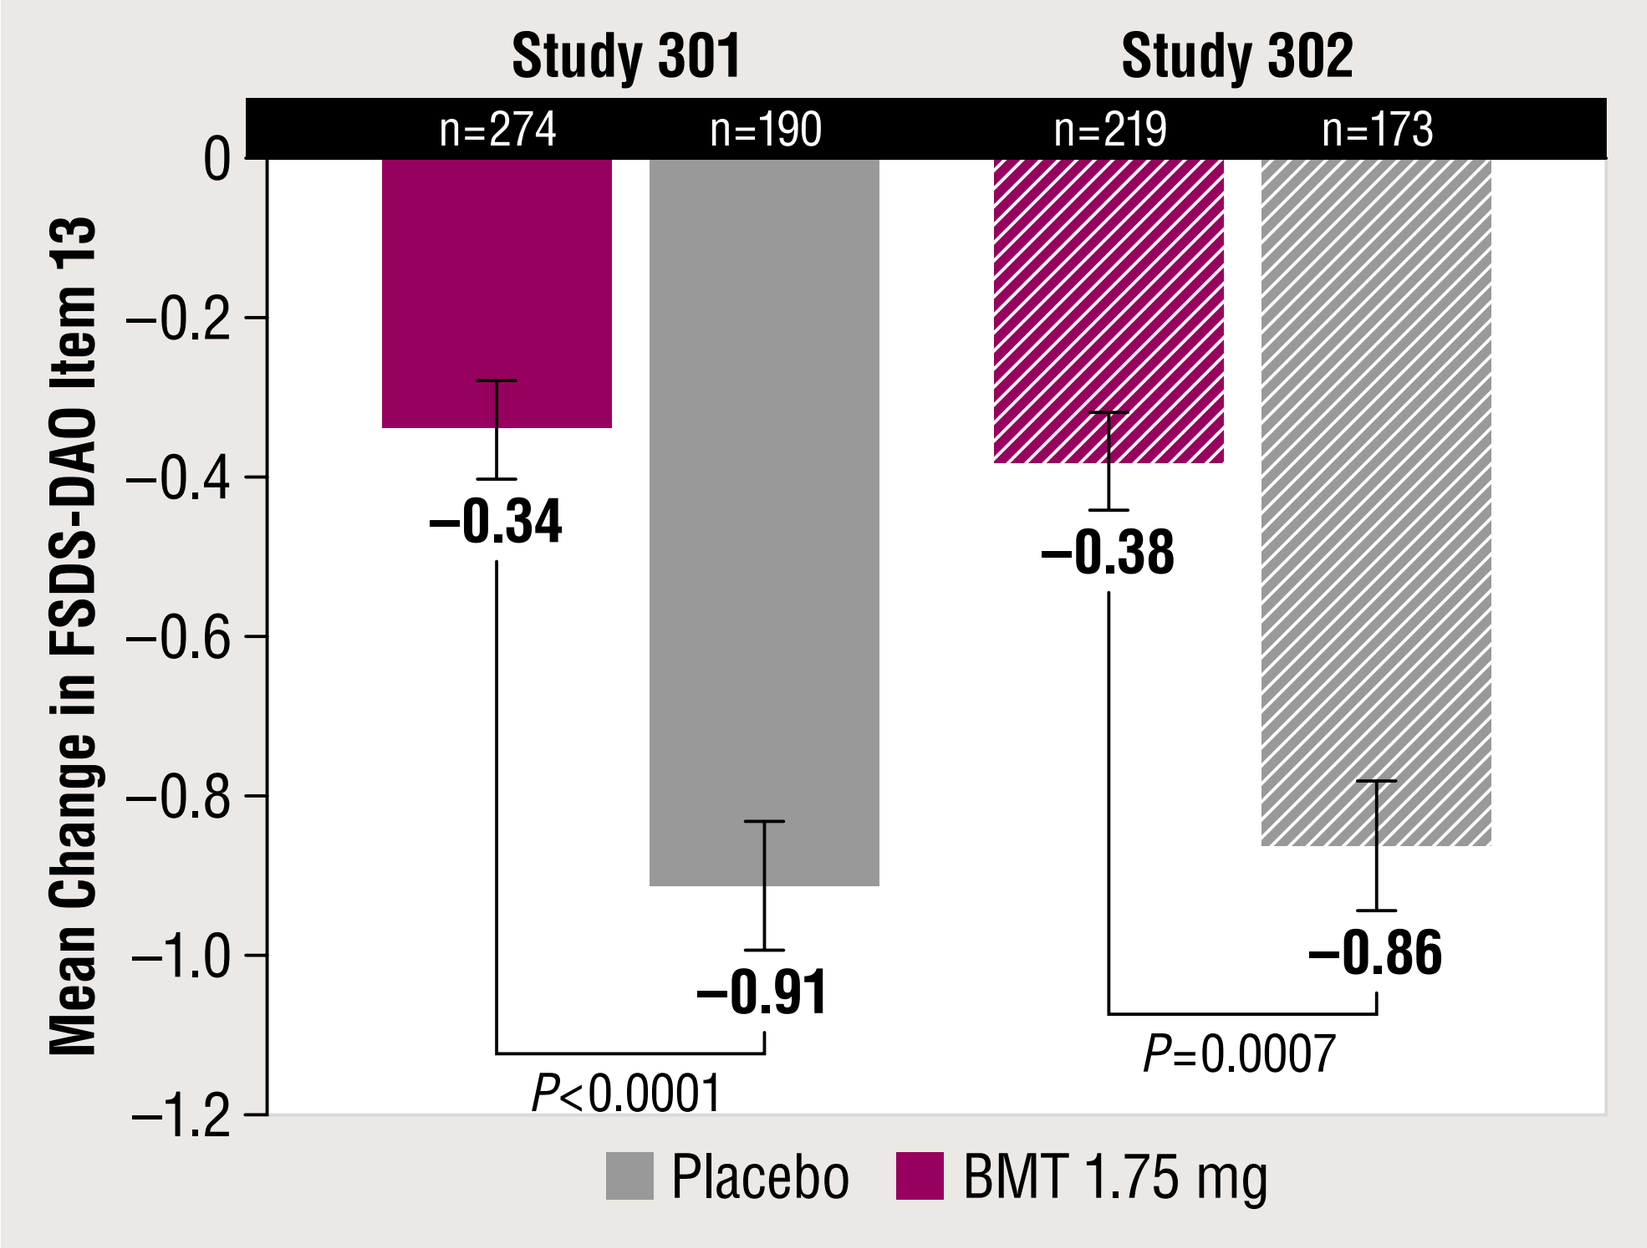

Study 301
Study 302
n=274
n=190
n=219
n=173
Mean Change in FSDS-DAO Item 13
0
−0.2
−0.4
−0.6
−0.8
−1.0
−1.2
−0.34
−0.91
P<0.0001
−0.38
−0.86
P=0.0007
Placebo
BMT 1.75 mg

Error bars are standard error of the mean.
*ie, "How often do you feel bothered by low sexual desire?" Scores range from 0 to 4 where 0 = never; 1 = rarely; 2 = occasionally; 3 = frequently; 4 = always.
BMT, bremelanotide; FSDS-DAO, Female Sexual Distress Scale–Desire/Arousal/Orgasm.
P-values determined by unadjusted Wilcoxon rank-sum test.

Responder Analyses (Table 2)

- 59% of women taking BMT in Study 301 and 58% in Study 302 were responders (self-assessed global benefit score ≥5 on Q3 of the GAQ), compared with 36% of women taking placebo in both studies
- The computed MCIDs were a median value of 0.6 for baseline to endpoint changes in FSFI-D subscale scores and a median 1.0-point improvement on FSDS-DAO Item 13
- Significant proportions of women taking BMT met the MCID thresholds for both coprimary endpoints in both studies

Table 2. Responder Analyses

% Responders Based on Self-Assessment of Benefit	Study 301		Study 302	
	PBO	BMT	PBO	BMT
GAQ Q3* score ≥5 (% responders BMT 1.75 mg SC)	36%	59%	36%	58%
Responder Analysis Based on MCIDs	**Study 301**		**Study 302**	
FSFI-D (MCID=0.6)	*P*=0.0002		*P*<0.0001	
FSDS-DAO Item 13 (MCID=−1.0)	*P*<0.0001		*P*=0.0419	

*ie, "To what degree do you think you benefited from taking the study drug?" (Answers: very much worse [1], no change [4], "very much better [7]).
BMT, bremelanotide; FSDS-DAO, Female Sexual Distress Scale-Desire/Arousal/Orgasm; FSFI-D, Female Sexual Function Index desire domain; GAQ, General Assessment Questionnaire; MCID, minimal clinically important difference; PBO, placebo; SC, subcutaneous.

Safety

- Most adverse events (AEs) were mild or moderate in nature
- Treatment-emergent AEs led to treatment discontinuation/interruption in approximately 18% of women taking bremelanotide (vs 2% in placebo)
- Most of the bremelanotide AEs causing withdrawal were nausea (9.9% of patients in Study 301 and 6.6% in Study 302) and vomiting (1.5% of patients in Study 301, and 0.7% in Study 302)
- Bremelanotide's safety profile was consistent with prior clinical trial experience; no new or unusual safety issues were identified for the double-blind period
- Bremelanotide has no known alcohol interaction[8]

References

1. Basson R. Women's sexual dysfunction: revised and expanded definitions. *CMAJ*. 2005;172:1327–1333. **2.** Shifren JL, Monz BU, Russo PA. Sexual problems and distress in United States women: prevalence and correlates. *Obstet Gynecol*. 2008;112:970–978. **3.** Molinoff PB, Shadiack AM, Earle D, Diamond LE, Quon CY. PT-141: a melanocortin agonist for the treatment of sexual dysfunction. *Ann N Y Acad Sci*. 2003;994:96–102. **4.** Pfaus J, Giuliano F, Gelez H. Bremelanotide: an overview of preclinical CNS effects on female sexual function. *J Sex Med*. 2007;4 Suppl 4:269–279. **5.** Meston CM. Validation of the Female Sexual 5. Function Index (FSFI) in women with female orgasmic disorder and in women with hypoactive sexual desire disorder. *J Sex Marital Ther*. 2003;29(1):39–46. **6.** Rosen R, Brown C, Heiman J, et al. The Female Sexual Function Index (FSFI): a multidimensional self-report instrument for the assessment of female sexual function. *J Sex Marital Ther*. 2000;26(2):191–208. **7.** Derogatis L, Clayton A, Lewis-D'Agostino D, Wunderlich G, Fu Y. Validation of the female sexual distress scale-revised for assessing distress in women with hypoactive sexual desire disorder. *J Sex Med.* 2008;5(2):357–364. **8.** Clayton AH, Lucas J, DeRogatis LR, Jordan R. Phase I Randomized Placebo-Controlled, Double-Blind Study of the Safety and Tolerability of Bremelanotide Coadministered With Ethanol in Healthy Male and Female Participants. *Clin Ther.* 2017;39:514–526.

Conclusions

- Treatment with BMT over the 6-month Core phase of the RECONNECT study was associated with clinically meaningful and statistically significant improvement in desire and a decrease in distress; both hallmark characteristics of HSDD
- FSFI-D scores showed that BMT significantly increased desire compared with placebo
- FSDS-DAO Item 13 scores in the BMT groups significantly decreased compared with placebo, indicating a decrease in distress related to low sexual desire
- The MCID analyses in the RECONNECT study, using a woman's own self-assessed benefit, provide support that the statistically significant numerical efficacy observed with BMT is clinically meaningful to women with HSDD

Support This study was funded by Palatin Technologies, Inc, Cranbury, New Jersey, USA. Editorial assistance was provided by The Curry Rockefeller Group, LLC, which was funded by Palatin Technologies, Inc.

Disclosures Sheryl Kingsberg has received research support or consulting fees from Palatin Technologies, Inc, Valeant, Novo Nordisk, Shionogi, Emotional Brain, Pfizer Inc, Trimel Pharmaceuticals, Inc (now Acerus), Nuelle, SST, TherapeuticsMD, Materna, Duquesney, Endoceutics, AMAG; and has stocks, stock options, or ownership interest, excluding diversified mutual funds, from Viveve. Robin Kroll has received research support or consulting fees from Bayer, Palatin Technologies, Inc, AbbVie, Shionogi, Trimel, Teva Pharmaceuticals, Activus, and Endoceutics. Johna Lucas, Robert Jordan, and Carl Spana are employees and stockholders of Palatin Technologies, Inc.